The Impact of Rheumatoid Arthritis on Patient-Reported Outcomes: Comparison Between Sarilumab Clinical Trials and Real-World Patient Data

Vibeke Strand,[1] Colleen M. Carpinella,[2] Lulu K. Lee,[2] Susan Boklage,[3] Matthew Reaney[4]

[1]Stanford University School of Medicine, Palo Alto, CA, USA; [2]Kantar Health, San Mateo, CA, USA; [3]Regeneron, Tarrytown, NY, USA; [4]Sanofi, Guildford, UK

ABSTRACT

Background/Purpose: Rheumatoid arthritis (RA) and its treatment have significant impacts on health-related quality of life (HRQoL), work productivity (WP) and activity participation. Data are limited comparing the magnitude of impacts observed in routine clinical practice and randomized controlled trials (RCTs). This study evaluated the impact of RA on HRQoL, WP and activity participation among adult RA patients in the US general population compared with patients from two sarilumab pivotal phase 3 RCTs. Sarilumab is a human anti-IL-6Rα monoclonal antibody for treatment of moderate-to-severely active RA.

Methods: "Real world" data were from adult participants of the cross-sectional 2015 US National Health and Wellness Survey (NHWS) with a patient-reported diagnosis of RA, moderate/severe disease activity and administered disease-modifying anti-rheumatic drugs (DMARDs) at the point of survey. RCT data were from MOBILITY – Part B and TARGET trials comparing the efficacy and safety of sarilumab subcutaneous (SC) 150 mg and 200 mg every 2 weeks (q2w) plus methotrexate in MOBILITY or conventional DMARDs in TARGET versus placebo in patients with moderate-to-severely active RA and inadequate responses to methotrexate (MTX-IR, MOBILITY) or tumor necrosis factors inhibitors (TNF-IR, TARGET). HRQoL was measured using the Short Form-36v2 (SF-36v2) in the NHWS, and both RCTs at baseline and Week 24; WP impairment (WPI) and activity impairment (AI) were measured via the WPAI scale in the NHWS and MOBILITY. Bivariate analyses evaluated between-group differences in SF-36v2 physical (PCS) and mental (MCS) component summary and domain scores and WPAI absenteeism, presenteeism (% work time missed), overall WPI and AI.

Results: Mean ages of the analyzed cohorts were 54, 51 and 53 years; 61%, 82%, and 80% were female, respectively in NHWS (n=2016), MOBILITY (n=799) and TARGET (n=365). At baseline, versus NHWS patients, those enrolled in MOBILITY reported greater presenteeism (47.25 vs 37.50; $P < 0.001$), overall WPI (51.29 vs 43.11; $P = 0.001$) and AI (62.42 vs 50.96; $P < 0.001$). Patients in MOBILITY and TARGET reported significantly lower baseline scores versus NHWS in SF-36v2 PCS (31.27, 29.42 vs 38.54), MCS (38.86, 38.73 vs 43.97), and all domain scores ($P < 0.001$) indicating greater HRQoL impairment. At Week 24 both trial cohorts reported improved HRQOL and AI in MOBILITY (34.08 vs 39.76, respectively; $P < 0.05$) following treatment with sarilumab than the NHWS cohort scores.

Conclusion: Patients in both sarilumab RCTs reported worse baseline HRQoL, and in MOBILITY greater WPI and AI than NHWS RA patients. After 24 weeks treatment with sarilumab, both RCT cohorts reported improvements exceeding NHWS scores, with few exceptions. These data offer perspective on the benefits of sarilumab treatment on HRQOL, WP and AI.

BACKGROUND

- The symptoms of rheumatoid arthritis (RA) are diverse and can have a detrimental effect on the health-related quality of life (HRQoL) of patients with the disease[1]
- HRQoL may be further impacted by the level of work productivity impairment (WPI) and activity impairment (AI) patients with RA may suffer
- Sarilumab is a human monoclonal antibody that blocks interleukin-6 (IL-6) from binding to both membrane-bound and soluble IL-6 receptor-α
 - It is approved for the treatment of moderately-to-severely active RA in adult patients who have had an inadequate response or intolerance to ≥1 disease-modifying antirheumatic drug (DMARD)
- Previous randomized controlled trials (RCTs) have demonstrated that sarilumab improves HRQoL, WPI and AI in patients with moderately-to-severely active RA;[2–4] however, it is not clear how these results compare with that of RA patients in the general population

OBJECTIVE

- To evaluate the impact of RA on HRQoL, WPI and AI among adult RA patients in the US general population compared with patients from two sarilumab pivotal phase 3 RCTs

METHODS

Data source

- Real-world data were derived from adult participants of the cross-sectional 2015 US National Health and Wellness Survey (NHWS)
- Participants were included if they:
 - had a patient-reported physician diagnosis of RA
 - had moderate-to-severe disease activity
 - were being administered biologic or conventional DMARDs at the point of survey
- RCT data were derived from patients with moderately-to-severely active RA participating in the MOBILITY and TARGET trials:
 - MOBILITY: sarilumab subcutaneous (SC) 150 mg or 200 mg every 2 weeks (q2w) plus methotrexate (MTX) in patients with inadequate response to MTX (MTX-IR)
 - TARGET: sarilumab SC 150 mg or 200 mg q2w plus conventional synthetic DMARDs in patients with inadequate response to tumor necrosis factor inhibitors (TNF-IR)

Measures

- HRQoL was measured using the Short Form-36 version 2 (SF-36v2) in NHWS at a single time point, and both RCTs at baseline and Week 24
- WPI and AI were measured using the Work Productivity and Activity Impairment (WPAI) scale in NHWS at a single time point and MOBILITY at baseline and Week 52
 - WPI and AI are not reported for TARGET as WPAI was not captured as part of the study

Statistical analysis

- Two-sample comparisons evaluated between-group differences in SF-36v2 physical and mental component (PCS and MCS, respectively) summary scores and individual domain scores, as well as WPAI absenteeism (days absent from work), presenteeism (impairment in productivity while working), overall WPI, and AI
- *P*-value represents nominal vs value linked to the proposed study hierarchy

RESULTS

- The study population included 2016 patients from NHWS, 799 from MOBILITY, and 365 from TARGET (Table 1)
 - Mean age was between 51 and 54 years
 - Approximately 80% of patients were female in the RCTs, but only 61% were female in NHWS ($P < 0.001$)

Table 1. Patient characteristics and demographics by RA cohort

	NHWS (N = 2016)	MOBILITY (N = 799)	TARGET (N = 365)
Age, mean years	53.9	50.5	53.4
Female, n (%)	1238 (61.4)	656 (82.1)	293 (80.3)
Non-Hispanic white, n (%)	1388 (68.8)	688 (86.1)	264 (72.3)
Duration of RA, mean years	13.5	9.0	12.1
Current medication, n (%)			
Biologic	388 (19.2)	161 (20.2)	343 (94.0)
DMARD	498 (24.7)	769 (96.2)	365 (100)
JAK	46 (2.3)	0	0

JAK, janus kinase inhibitor

- Patients enrolled in MOBILITY had greater absenteeism, presenteeism, overall WPI, and AI at baseline compared with patients from NHWS (Figure 1)
- Following 52 weeks of treatment, patients in MOBILITY reported improvements in WPI, AI, absenteeism and presenteeism versus baseline,[4] resulting in less absenteeism, presenteeism, WPI and AI than patients from NHWS (Figure 1)
- Patients in MOBILITY and TARGET reported lower (worse) baseline component summary scores for SF-36v2 PCS and MCS compared with patients from NHWS:
 - PCS: 31.3 for MOBILITY, 29.4 for TARGET, and 38.5 for NHWS ($P < 0.001$ for both MOBILITY and TARGET vs NHWS)
 - MCS: 38.9 for MOBILITY, 38.7 for TARGET, and 44.0 for NHWS ($P < 0.001$ for both MOBILITY and TARGET vs NHWS)
- Following 24 weeks of treatment with sarilumab, patients in MOBILITY and TARGET reported improvements in SF36v2 PCS (MOBILITY: 39.5; TARGET 39.0) and MCS (MOBILITY: 45.1; TARGET: 46.4) scores compared with baseline. Furthermore, these scores were greater than those seen in NHWS patients (PCS: 44.0; MCS: 38.5)

Figure 1. Work and activity impairment in MOBILITY and NHWS patients

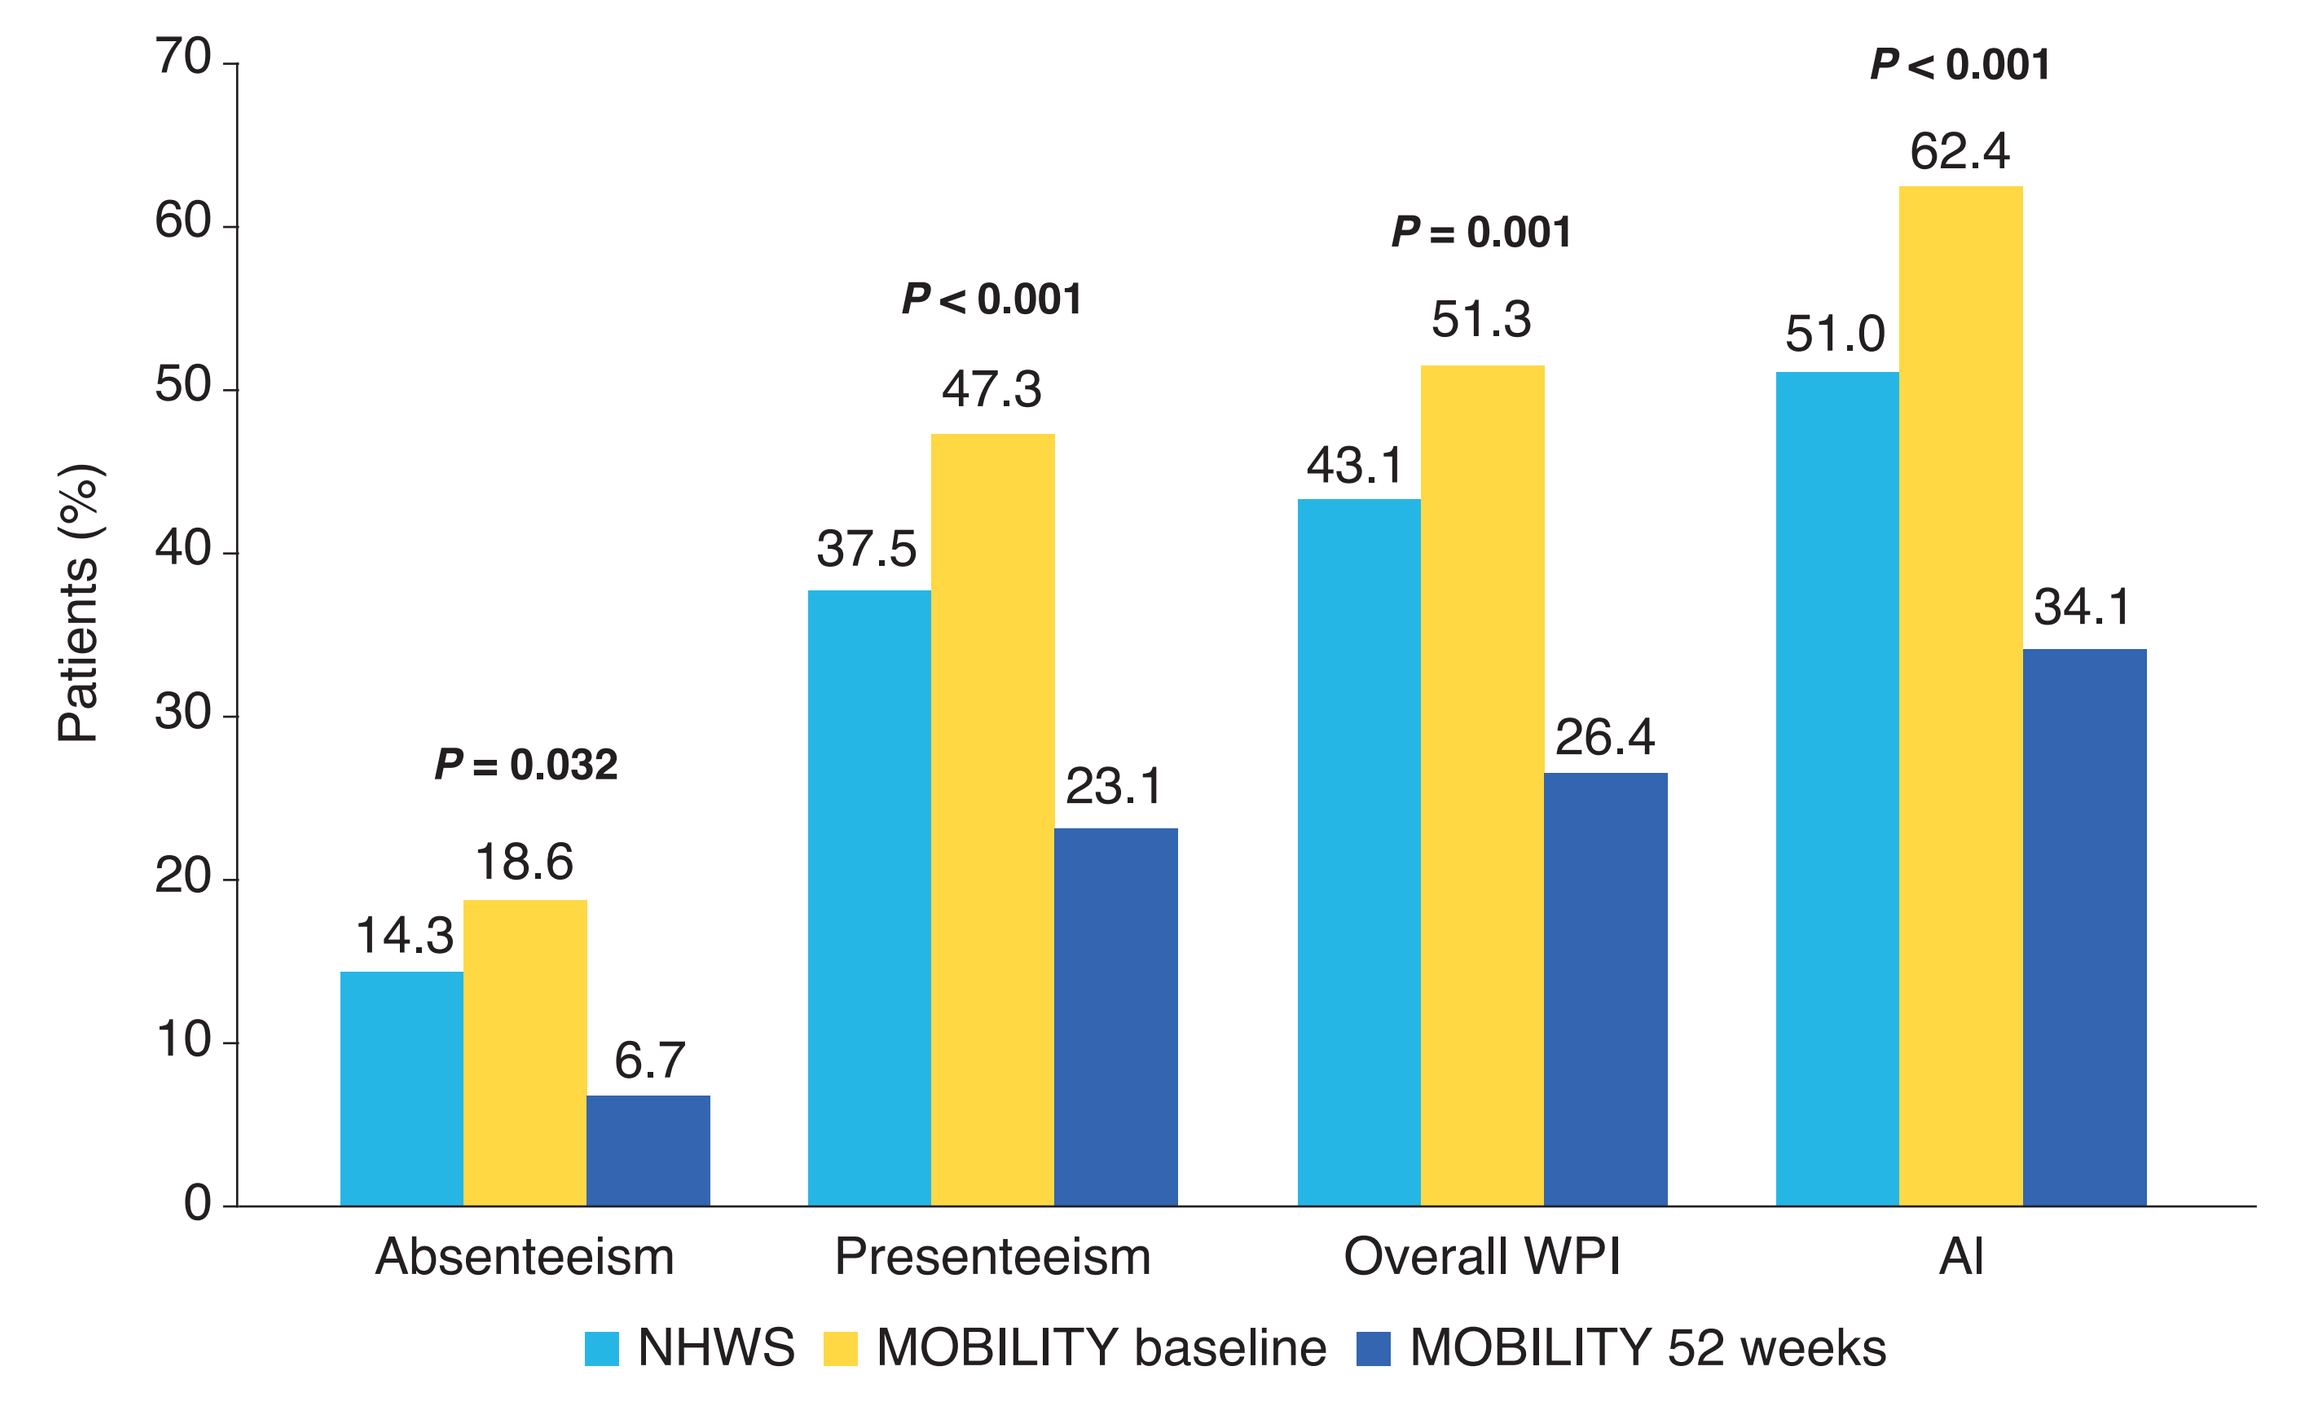
Patients (%)
70
60
50
40
30
20
10
0
P = 0.032
14.3
18.6
6.7
P < 0.001
37.5
47.3
23.1
P = 0.001
43.1
51.3
26.4
P < 0.001
51.0
62.4
34.1
Absenteeism
Presenteeism
Overall WPI
AI
NHWS
MOBILITY baseline
MOBILITY 52 weeks

P-values are MOBILITY baseline versus NHWS

- Patients in MOBILITY and TARGET reported significantly lower SF-36v2 domain scores at baseline versus NHWS patients ($P < 0.001$), indicating greater HRQoL impairment (Figure 2)
- Following 24 weeks of treatment with sarilumab in the RCTs, patients reported improvements in HRQoL demonstrated by higher SF-36v2 domain scores versus baseline. Furthermore, these scores were greater than those seen in NHWS patients (Figure 2)

Figure 2. SF-36v2 individual domain scores for NHWS versus MOBILITY and TARGET

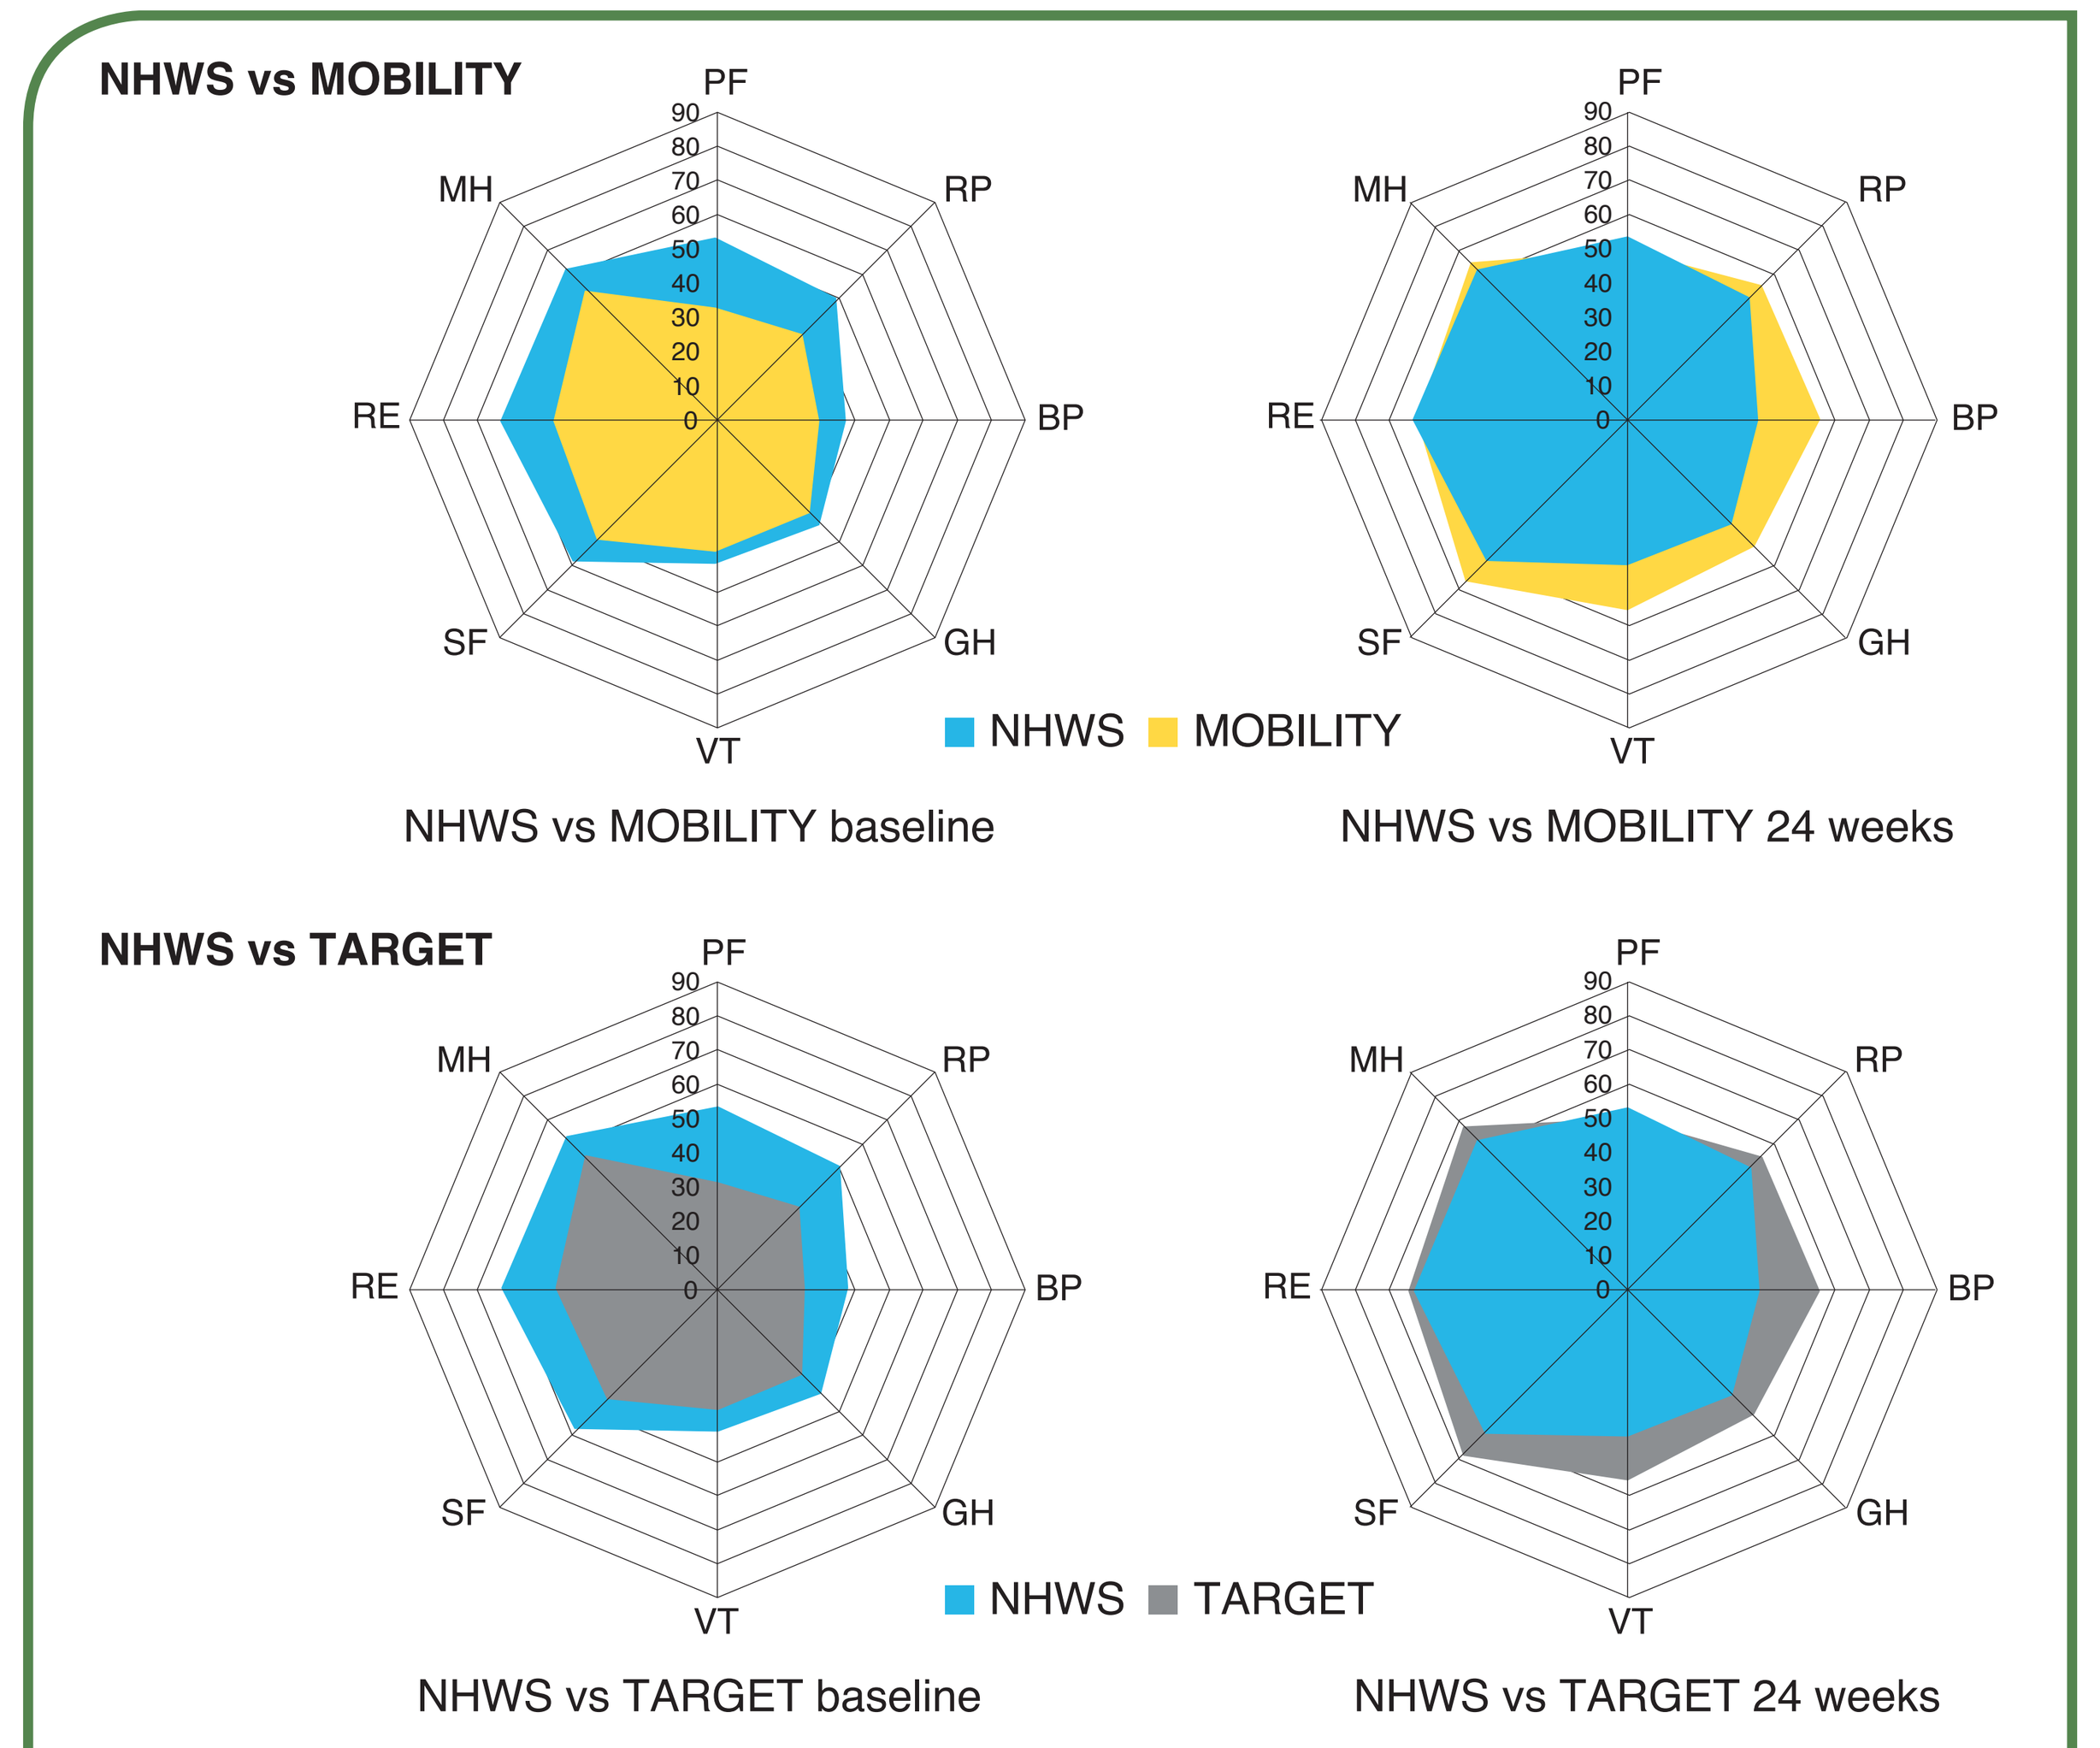
NHWS vs MOBILITY
PF
RP
BP
GH
VT
SF
RE
MH
NHWS
MOBILITY
NHWS vs MOBILITY baseline
NHWS vs MOBILITY 24 weeks
NHWS vs TARGET
NHWS
TARGET
NHWS vs TARGET baseline
NHWS vs TARGET 24 weeks

BP, bodily pain; GH, general health; MH, mental health; PF, physical functioning; RE, role emotional; RP, role physical; SF, social functioning; VT, vitality.
NHWS vs MOBILITY baseline: all *P*-values < 0.001. NHWS vs MOBILITY 24 weeks: all *P*-values < 0.01, except for RE ($P = 0.425$). NHWS vs TARGET baseline: all *P*-values < 0.001. NHWS vs TARGET 24 weeks: all *P*-values < 0.01, except for PF ($P = 0.105$), RP ($P = 0.015$) and RE ($P = 0.529$).

SAFETY

- Sarilumab safety was consistent with IL-6 receptor blockade
- The two most common adverse events were infection and neutropenia

CONCLUSIONS

- Patients with moderate-to-severely active RA in two sarilumab RCTs reported worse baseline HRQoL than similar patients in routine clinical practice (NHWS)
- However, over the course of the two RCTs, patients receiving sarilumab 150 mg or 200 mg showed improvements in SF-36 domain scores, WPI and AI with better scores than those in routine clinical practice (NHWS)

References
1. Matcham F et al. *Semin Arthritis Rheum*. 2014;44:123–130; 2. Burmester G et al. *Ann Rheum Dis*. 2017;76:840–847; 3. Strand V et al. *RMD Open*. 2017;3:e000416; 4. Strand V et al. *Arthritis Res Ther*. 2016;18:198.

Acknowledgements
The study was funded by Sanofi and Regeneron Pharmaceuticals, Inc. Medical writing support was provided by Kerren Davenport, Prime, Knutsford, Cheshire, UK.

Disclosures
VS has received consulting fees from AbbVie, Amgen, AstraZeneca, Bayer, Boehringer Ingelheim, Celltrion, Corrona, Crescendo Bioscience, Eli Lilly, Genentech/Roche, GlaxoSmithKline, Horizon, Janssen, Merck, Novartis, Pfizer, Regeneron Pharmaceuticals, Inc., Samsung, Sandoz, Sanofi, and UCB. CC and LL are employees of Kantar Health, a company that has received funding to conduct this study. SB is an employee of and stockholder in Regeneron Pharmaceuticals, Inc. MR is an employee of and stockholder in Sanofi.

Presented at the ACR/ARHP Annual Meeting, October 19–24, 2018, Chicago, IL, USA